Poster# **R106**

Panagiotis Orfanos | panagiotis.orfanos@novartis.com

CLINICAL RELEVANCE OF URTICARIA ACTIVITY SCORES FROM THE PATIENT PERSPECTIVE

Diane Whalley[1]; Sarah Sauchelli[1]; Maria-Magdalena Balp[2]; Ravneet Kohli[3]; Panagiotis Orfanos[2]

[1]RTI Health Solutions, Manchester, United Kingdom; [2]Novartis Pharma AG, Basel, Switzerland; [3]Novartis Healthcare Private Limited, Hyderabad, India

KEY FINDINGS & CONCLUSIONS

- Compared with UAS7 scores of 16 or more (moderate or severe urticaria activity categories), UAS7 scores of 6 or below (urticaria free or well controlled urticaria categories) were associated with meaningfully better status on patient reported measures and patient experiences of symptom severity, disease control, health-related quality of life (HRQOL), sleep, daily activities, and rescue medication use
- While the complete resolution of symptoms (UAS7 = 0) with treatment is optimal, the achievement of UAS7 scores of 1 to 6 is a meaningful and clinically relevant outcome from the patient perspective

Scan to obtain:
- Poster
- Plain language summary
- Slides
- Supplementary material

https://bit.ly/ACCAI-R106

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

This study is sponsored by Novartis Pharma AG.
Poster presented at the ACAAI 2024, held on 24-28 October 2024; Boston, MA, United States

INTRODUCTION

- The Urticaria Activity Score (UAS) is a diary-based patient-reported outcome (PRO) measure assessing daily severity of itch and number of hives. Weekly scores from the measure (UAS7) are widely used to assess disease activity in chronic spontaneous urticaria (CSU) clinical trials
- Understanding the meaningfulness of UAS7 scores from the patient perspective is important to determine their clinical relevance
- The study objective was to determine the clinical relevance of UAS7 scores that are often used to define moderate-to-severe disease activity as a criterion for enrolment in clinical trials (UAS7 ≥ 16) and to define a meaningful treatment response (UAS7 ≤ 6)

METHODS

- Analyses were conducted using an initial data cut from two phase III clinical trials of remibrutinib in adults with CSU inadequately controlled by H1-antihistamines (REMIX 1 [NCT05030311] and REMIX 2 [NCT05032157]). Data at baseline and week 12, pooled across treatment arms and studies, were analyzed
 - The UAS was administered as part of the Urticaria Patient Daily Diary (UPDD)[1,2]
 - UAS7 scores range from 0 to 42, where higher scores indicate more severe itch and hives
 - Score categories were defined as follows:[3] UAS7 = 0, urticaria free; UAS7 > 0 to 6, well-controlled disease activity; UAS7 > 6 to < 16, mild activity; UAS7 = 16 to < 28, moderate activity; and UAS7 = 28 to 42 = severe activity
 - Additional study measures included in the analyses: Dermatology Life Quality Index (DLQI); Urticaria Control Test 7-day recall version (UCT7); Patient Global Impression of Severity (PGIS), weekly Activity Interference Score (AIS7), weekly Sleep Interference Score (SIS7), and number of rescue medication tablets taken over 7 days
 - Analysis of variance or chi-squared test were used to evaluate differences in scores on the other study measures across the UAS7 score categories
- Qualitative interviews were conducted as part of a standalone study with adults in the United States with CSU who were symptomatic and taking H1-antihistamines
 - Participants were presented with different scenarios depicting different UAS7 scores and asked to provide feedback on the symptoms and their impact that would be associated with each score
 - Deductive content analysis methods were applied to examine patients' perspective on the meaningfulness of the UAS7 score categories

RESULTS

Analysis of REMIX-1 and REMIX-2 Data

Sample Characteristics and Descriptive Statistics

- The sample included 889 participants in the REMIX I and REMIX II full analysis sets with UAS data at baseline and weeks 4 or 12 at the time the data were drawn for analysis
- Mean (standard deviation [SD]) age was 43.4 (14.27) years, 66.3% were female, 55.5% were White, and 83.9% identified as not Hispanic or Latino
- Mean (SD) UAS7 improved from 30.32 (7.77) at baseline to 11.88 (11.47) at week 12 (**Table 1**)

Table 1. Descriptive Statistics for the UAS7 Scores

Parameter	Baseline	Week 12	Change from baseline to week 12
n	889	743	743
Mean (SD)	30.32 (7.77)	11.88 (11.47)	−18.50 (12.56)
Median (Q1, Q3)	30.00 (24.00, 37.00)	10.00 (1.00, 19.50)	−19.50 (−28.00, −9.00)
Minimum, maximum	6.5, 42.0	0.0, 42.0	−42.0, 15.0

SD = standard deviation; Q = quartile; UAS7 = weekly Urticaria Activity Score.

- Most participants (99.1%) had moderate or severe urticaria activity at baseline (**Table 2**). At week 12, 41.0% of participants were urticaria free or had well controlled urticaria (UAS7 ≤ 6) and 30.8% had moderate or severe urticaria activity (UAS7 ≥ 16) (**Table 2**)

Table 2. UAS7 categories scores at baseline and week 12

UAS7 score categories	Baseline, n (%)	Week 12, n (%)
Urticaria free (UAS7 = 0)	0 (0.0)	174 (23.4)
Well-controlled (UAS7 > 0 to ≤ 6)	0 (0.0)	131 (17.6)
Mild activity (UAS7 > 6 to < 16)	8 (0.9)	209 (28.1)
Moderate activity (UAS7 ≥ 16 to < 28)	327 (36.8)	124 (16.7)
Severe activity (UAS7 ≥ 28 to ≤ 42)	554 (62.3)	105 (14.1)
Missing	0 (0.0)	146 (16.4)

UAS7 = weekly Urticaria Activity Score; Note: Percentages exclude missing data.

DLQI and UCT7 Scores Across UAS7 Score Categories at Week 12

- Descriptive statistics for DLQI and UCT7 scores by UAS7 score categories are presented in **Table 3**
- Total scores on the DLQI and UCT7 were significantly different between the UAS7 score categories at week 12 (all P values < 0.0001; **Figure 1**)
- Participants categorized as urticaria free or well controlled urticaria (UAS ≤ 6) had median DLQI scores indicating no effect on HRQOL (DLQI 0 or 1)[4], whereas participants categorized as moderate or severe urticaria activity (UAS7 ≥ 16) had median DLQI scores indicating moderate to very large effect on life (DLQI ≥ 6)
- Participants categorized as urticaria free or well controlled urticaria (UAS ≤ 6) had median UCT7 scores indicating complete control or well-controlled disease (UCT7 ≥ 12)[5,6], whereas participants categorized as moderate or severe activity (UAS7 ≥ 16) had median UCT7 scores indicating disease that is not well controlled (UCT7 < 12)

Table 3. Descriptive Statistics for DLQI and UCT7 Scores by UAS7 Score Categories

Supporting measure	UAS7 score category				
	Urticaria free (0)	Well-controlled (> 0 to 6)	Mild activity (> 6 to < 16)	Moderate activity (16 to < 28)	Severe activity (28 to 42)
DLQI total score					
Mean (SD), n	0.61 (1.41), 160	2.83 (3.61), 128	5.91 (4.69), 197	8.80 (5.87), 120	12.35 (6.99), 101
Median (Q1, Q3)	0.0 (0.0, 0.0)	1.0 (1.0, 4.0)	5.0 (2.0, 9.0)	7.5 (4.0, 12.0)	11.0 (8.0, 17.0)
95% CI	0.4, 0.8	2.2, 3.5	5.3, 6.6	7.7, 9.9	11.0, 13.7
UCT7 total score					
Mean (SD), n	15.05 (1.81), 160	12.75 (2.75), 128	10.31 (3.08), 197	7.98 (2.77), 120	6.04 (3.62), 100
Median (Q1, Q3)	16.0 (15.0, 16.0)	13.0 (12.0, 15.0)	11.0 (8.0, 12.0)	8.0 (6.0, 9.5)	6.0 (3.0, 8.0)
95% CI	14.8, 15.3	12.3, 13.2	9.9, 10.7	7.5, 8.5	5.3, 6.8

ANOVA = analysis of variance; CI = confidence interval; DLQI = Dermatology Life Quality Index; Q = quartile; SD = standard deviation; UAS7 = weekly Urticaria Activity Score; UCT7 = Urticaria Control Test 7-day recall version. Note: ANOVA P values all < 0.0001.

Patient-Reported Symptom Severity Across UAS7 Score Categories at Week 12

- At week 12, participants categorized as urticaria free (UAS7 = 0) or well-controlled urticaria (UAS7 > 0 to 6) were most likely to rate their symptoms on the PGIS as none or mild, whereas participants categorized as moderate (UAS7 = 16 to < 28) or severe (UAS7 = 28-42) were most likely to rate their symptoms as moderate to very severe (**Table 4**; P < 0.0001)

Table 4. Frequencies and Percentages of PGIS Responses by UAS7 Score Categories at Week 12

PGIS response	UAS7 score category, n (%)				
	Urticaria free (0)	Well-controlled (> 0 to 6)	Mild activity (> 6 to < 16)	Moderate activity (16 to < 28)	Severe activity (28 to 42)
No symptoms	131 (82.4)	37 (28.9)	10 (5.1)	1 (0.8)	0 (0.0)
Mild	25 (15.7)	77 (60.2)	122 (61.9)	27 (22.5)	12 (11.9)
Moderate	2 (1.3)	11 (8.6)	53 (26.9)	70 (58.3)	44 (43.6)
Severe	1 (0.6)	3 (2.3)	12 (6.1)	20 (16.7)	27 (26.7)
Very severe	0 (0.0)	0 (0.0)	0 (0.0)	2 (1.7)	18 (17.8)

PGIS = Patient Global Impression of Severity; UAS7 = weekly Urticaria Activity Score. Note: X^2 P value < 0.0001

Activity and Sleep Interference, Angioedema, and Usage of Rescue Medication Across UAS7 Score Categories at Week 12

- Scores on the AIS7, SIS7, number of tablets of rescue medication taken over 7 days, and number of days with angioedema over 7 days were significantly different between the UAS7 score categories at week 12 (all P values < 0.0001; **Table 5**)
- Participants categorized as urticaria free or well controlled urticaria (UAS ≤ 6) had median AIS7 and SIS7 scores indicating no interference (AIS7 and SIS7 = 0), whereas participants categorized as moderate or severe activity (UAS7 ≥ 16) had higher median scores (7.0 to 14.0; **Table 5**)
- Participants categorized as urticaria free or well controlled urticaria (UAS ≤ 6) had a median of 0 tablets of rescue medication over 7 days, whereas participants categorized as moderate or severe activity (UAS7 ≥ 16) had higher median numbers of tablets (3.0 to 8.0; **Table 5**)

Figure 1. DLQI and UCT7 Scores by UAS7 Score Categories at Week 12

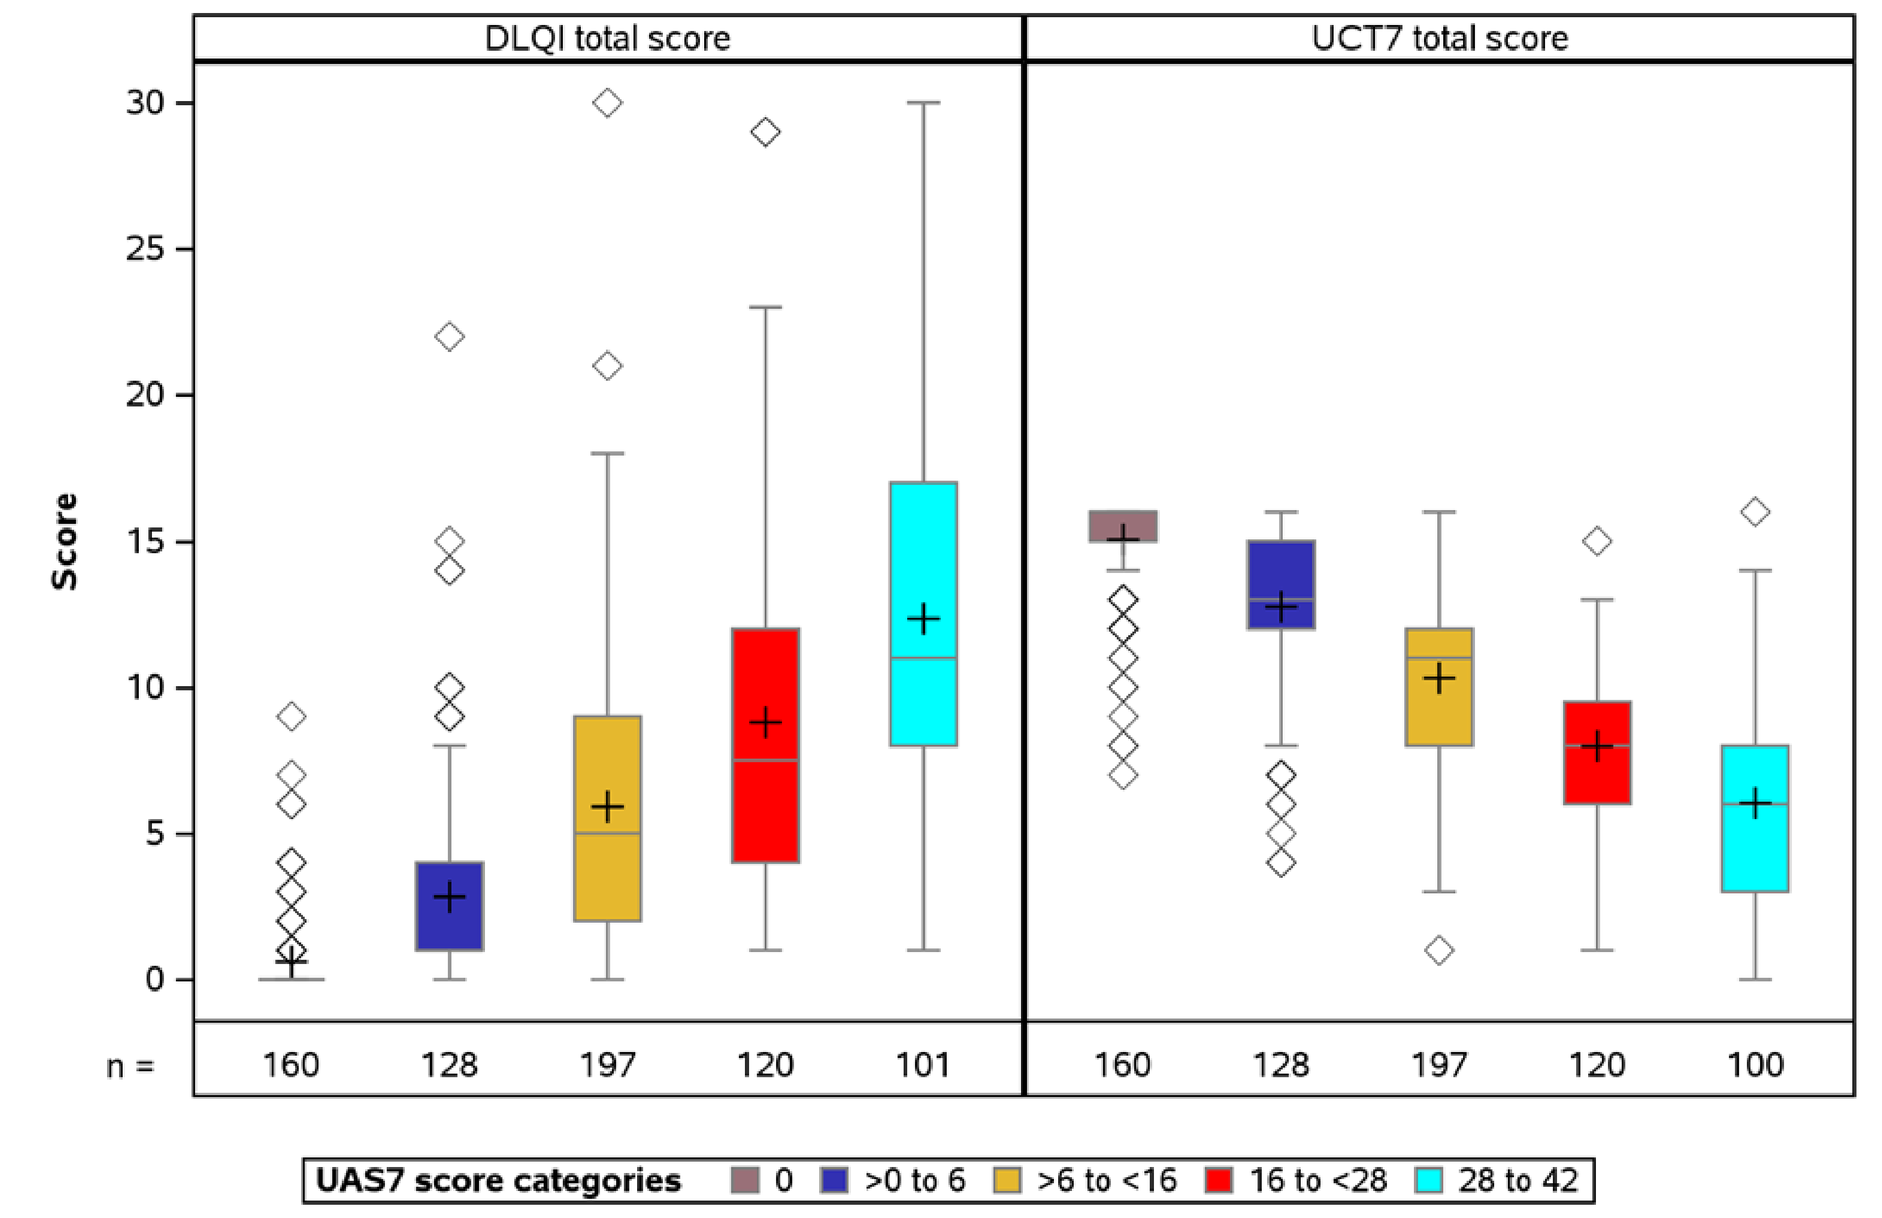
DLQI total score
UCT7 total score
Score
30
25
20
15
10
5
0
n =
160
128
197
120
101
160
128
197
120
100
UAS7 score categories
0
>0 to 6
>6 to <16
16 to <28
28 to 42

DLQI = Dermatology Life Quality Index; UAS = Urticaria Activity Score; UAS7 = weekly Urticaria Activity Score; UCT7 = Urticaria Control Test 7-day recall version.

Table 5. Descriptive Statistics for Weekly Activity Interference and Sleep Interference Scores and Rescue Medication Use by UAS7 Score Categories

Supporting measure	UAS7 score category				
	Urticaria free (0)	Well-controlled (> 0 to 6)	Mild activity (> 6 to < 16)	Moderate activity (16 to < 28)	Severe activity (28 to 42)
AIS7					
Mean (SD), n	0.00 (0.00), 169	0.89 (1.44), 126	4.39 (3.14), 203	7.92 (3.76), 121	13.42 (4.85), 102
Median (Q1, Q3)	0.0 (0.0, 0.0)	0.0 (0.0, 1.2)	5.0 (1.2, 7.0)	8.0 (6.0, 10.5)	14.0 (11.0, 16.0)
95% CI	–	0.6, 1.1	4.0, 4.8	7.2, 8.6	12.5, 14.4
SIS7					
Mean (SD), n	0.00 (0.00), 168	0.79 (1.60), 126	3.60 (3.14), 201	7.04 (4.06), 120	13.50 (5.42), 102
Median (Q1, Q3)	0.0 (0.0, 0.0)	0.0 (0.0, 1.0)	3.0 (0.0, 7.0)	7.0 (4.0, 10.0)	14.0 (11.2, 16.0)
95% CI	–	0.5, 1.1	3.2, 4.0	6.3, 7.8	12.4, 14.6
Number of tablets of rescue medication over 7 days					
Mean (SD), n	3.38 (6.72), 88	3.79 (6.84), 71	7.41 (11.39), 107	7.19 (8.02), 70	12.93 (13.97), 57
Median (Q1, Q3)	0.0 (0.0, 6.0)	0.0 (0.0, 6.0)	3.0 (0.0, 12.0)	4.0 (0.0, 12.0)	8.0 (2.0, 20.0)
95% CI	2.0, 4.8	2.2, 5.4	5.2, 9.6	5.3, 9.1	9.2, 16.6

AIS7 = weekly Activity Interference Score; ANOVA = analysis of variance; CI = confidence interval; Q = quartile; SD = standard deviation; SIS7 = weekly Sleep Interference Score; UAS7 = weekly Urticaria Activity Score. Note: ANOVA P values all < 0.0001.

Patients' Perspective on the Meaningfulness of UAS7 Scores

- Qualitative interviews were conducted with 15 adults with CSU
- The mean (SD) age of the interview sample was 42.7 (13.1) years, 86.7% were female, 80.0% identified as White, and 73.3% were in full-time employment
- Interview participants generally considered that a UAS7 score of 6 represented a desirable outcome, with symptoms that were well-controlled by treatment and having meaningful benefits in many aspects of their daily life
- In contrast, the UAS7 score of 16 was considered to represent meaningfully worse urticaria than UAS7 scores of 11 and/or 6, in terms of rescue medication use, discomfort experienced, and disruption to their sleep, daily activities, and emotional well-being
- Overall, participants indicated they would consider changing their treatment if they were experiencing symptoms associated with a UAS7 score of 16, and that they would be satisfied with the level of symptoms associated with a UAS7 score of 6

References

1.Mathias SD et al. Ann Allergy Asthma Immunol. 2010 Aug;105(2):142-8. **2.** Mathias SD et al. Allergy Asthma Proc. 2012 ;33(2):186-90; **3.** Stull D et al. Br J Dermatol. 2017 Oct;177(4):1093-101. **4.**Hongbo Y et al.J Invest Dermatol. 2005 Oct;125(4):659-64 **5.**Buttgereit T, et al.J Allergy Clin Immunol. 2023 May 18:S0091-6749(23)00604-8. **6.**Weller K, et al. J Allergy Clin Immunol. 2014 May;133(5):1365-72, 1372.e1-6.

Acknowledgements

Editorial and medical writing support was provided by Samprati Avasthi (Novartis Healthcare Pvt. Ltd., Hyderabad, India) in accordance with Good Publication Practice (GPP3) guidelines(www.ismpp.org/gpp3). The final responsibility for the content lies with the authors

Disclosures

This study was funded by Novartis Pharma AG, Basel, Switzerland.