009

Alexander Greiner I alexgreiner@allergyandasthma.com

Remibrutinib treatment has no impact on mean blood cell counts in patients with chronic spontaneous urticaria: Results from Phase 2, 52-week extension study

Alexander Greiner,[1] Vipul Jain,[2] Ana M Giménez-Arnau,[3] Jeffrey Tillinghast,[4] Karine Lheritier,[5] Pauline Walsh,[6] Sibylle Haemmerle,[5] Artem Zharkov,[5] Robert Snyder[7]

[1]Allergy and Asthma Medical Group and Research Center, and University of California, San Diego; [2]Division of Clinical Immunology and Allergy, Department of Medicine, McMaster University, Hamilton, Canada; [3]Department of Dermatology, Hospital del Mar Research Institute, Universitat Pompeu Fabra, Barcelona, Spain; [4]The Clinical Research Center, St. Louis, Missouri, USA; [5]Novartis Pharma AG, Basel, Switzerland; [6]Novartis Ireland Limited, Dublin, Ireland; [7]Riverchase Dermatology, Pembroke Pines, Florida, USA.

Scan to obtain:
• Poster

To download a copy of this poster, visit the web at:
https://bit.ly/UKCAAAAI

Copies of this poster obtained through quick response (QR) code are for personal use only and may not be reproduced without written permission of the author

CONCLUSIONS

- Overall, during the Phase 2 extension study, remibrutinib treatment did not impact the mean blood cell count
- These outcomes are in line with the remibrutinib safety profile observed in the Phase 2 core and extension studies[3,4]
- Data from REMIX-1 and REMIX-2 Phase 3 studies will provide more insights on the safety profile of remibrutinib

Poster presented at: The American Academy of Allergy, Asthma & Immunology Annual Meeting; February 23-26, 2024; Washington, DC.

INTRODUCTION

- Chronic spontaneous urticaria (CSU) is characterized by occurrence of hives (itchy wheals) and/or angioedema for more than 6 weeks and can have a major impact on patients' well-being[1]
- Remibrutinib is a novel, oral, highly selective Bruton's tyrosine kinase (BTK) inhibitor,[2] that has demonstrated safety and efficacy for up to 52 weeks in the Phase 2 core and extension studies (NCT03926611 and NCT04109313)[3,4] and in the 24 weeks primary analysis of both the Phase 3 studies (REMIX-1: NCT05030311, REMIX-2: NCT05032157) in patients with CSU inadequately controlled by H1-antihistamines (H1-AH)[5]
- BTK is expressed in hematopoietic cells including myeloid cells.[6] This analysis aimed to examine the impact of remibrutinib treatment on complete blood count parameters

OBJECTIVE

- To evaluate the effect of remibrutinib versus placebo with respect to change from baseline (CFB) in blood cell counts from the Phase 2, 52-week extension study in patients with CSU

METHODS

Study design

- This was an open-label, single-arm, multicenter, long-term Phase 2 extension study conducted across 15 countries in patients with CSU inadequately controlled by H1-AH (**Figure 1**)[4]
- Eligible patients (weekly Urticaria Activity Score [UAS7] ≥16 at the end of the treatment or follow-up period of the Phase 2 core study, or by the end of the observational period) were enrolled in the extension[4]
- Enrolled patients received remibrutinib 100 mg twice daily (b.i.d.)[4]

Study assessments and analysis

- Mean (standard deviation [SD]) values at baseline and CFB in blood cell count (for leukocytes, neutrophils, eosinophils, basophils, lymphocytes, erythrocytes, and platelets) were assessed up to Week 52
- All analyses were performed at a descriptive level and the data are presented as observed

Figure 1. Study design[4]

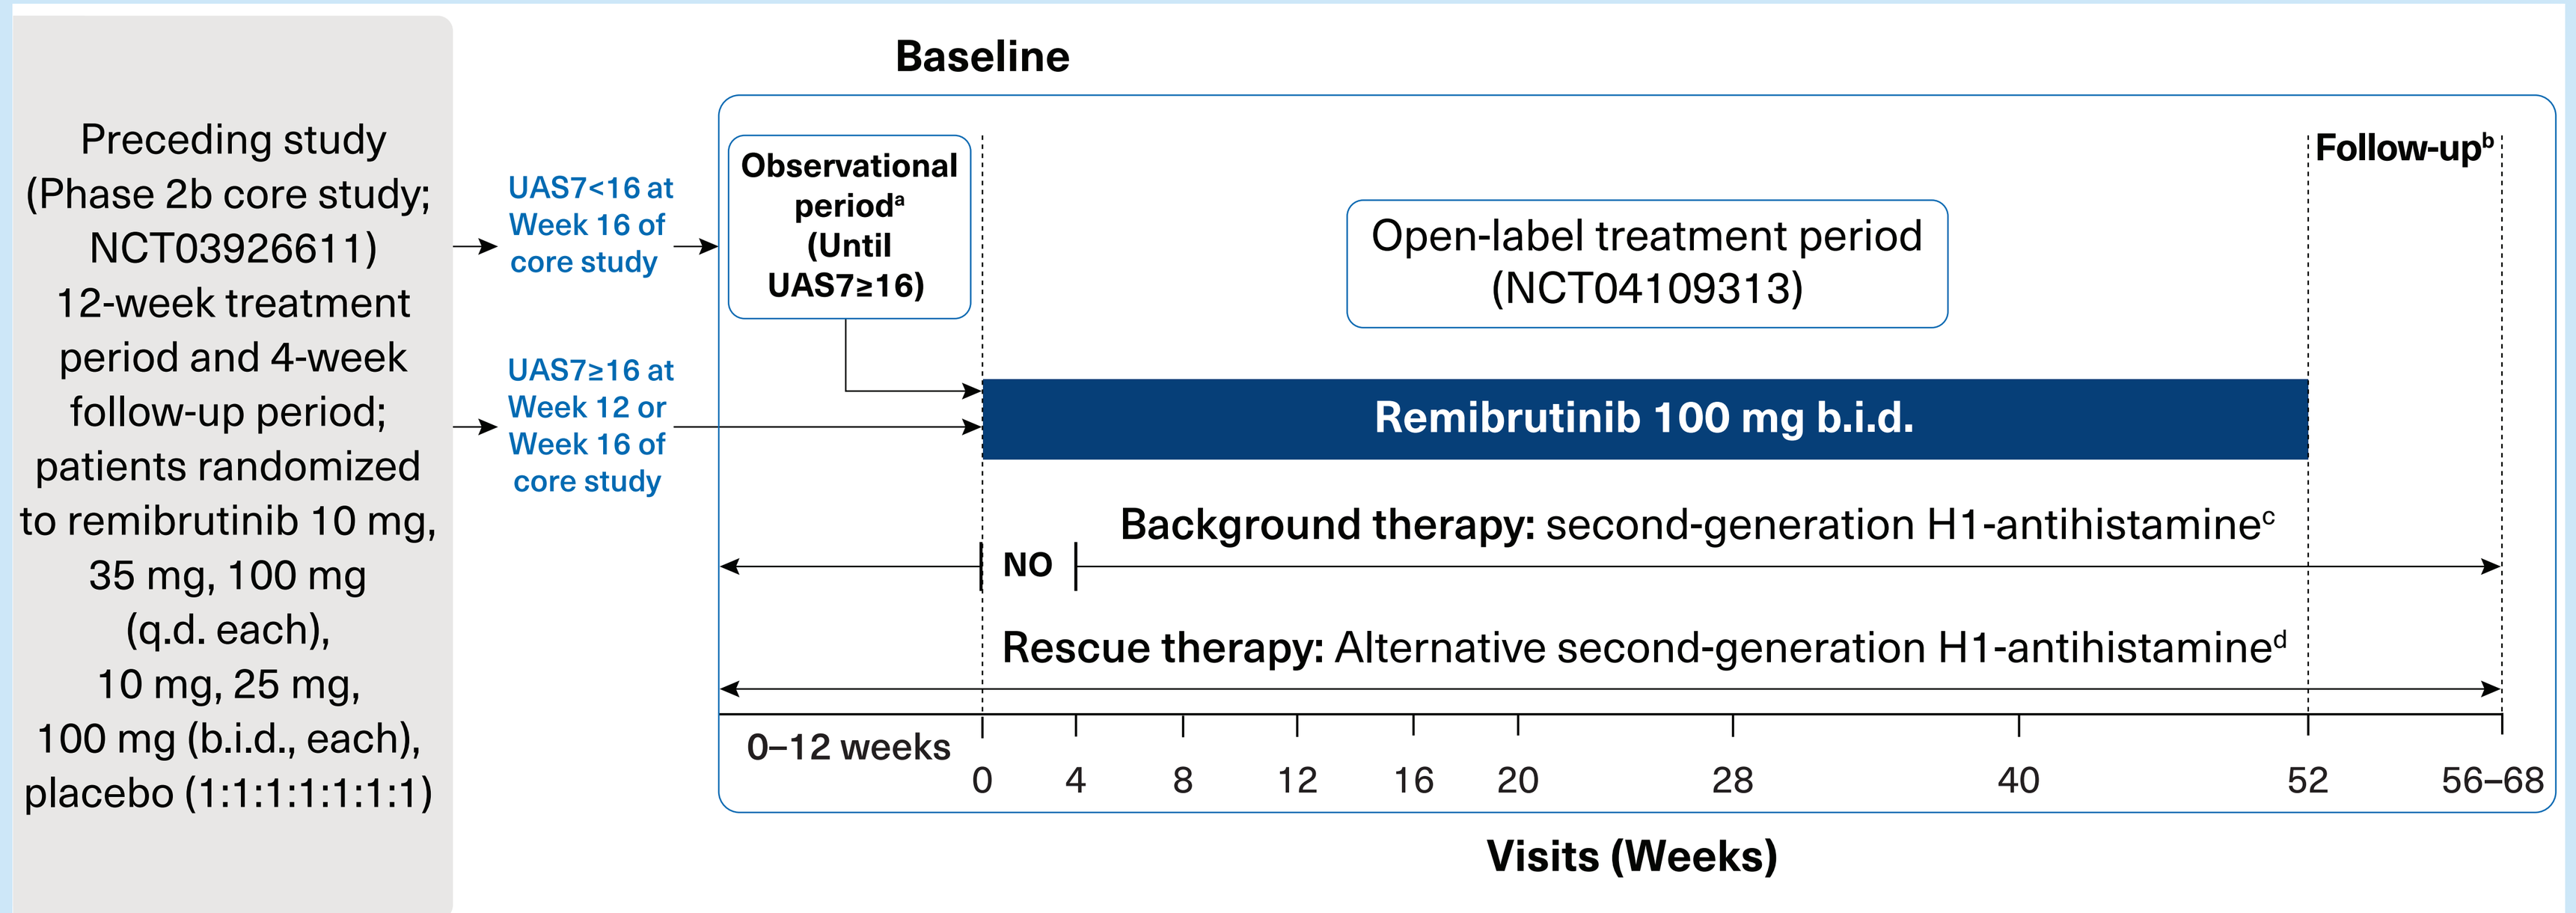
Preceding study (Phase 2b core study; NCT03926611) 12-week treatment period and 4-week follow-up period; patients randomized to remibrutinib 10 mg, 35 mg, 100 mg (q.d. each), 10 mg, 25 mg, 100 mg (b.i.d., each), placebo (1:1:1:1:1:1:1)
UAS7<16 at Week 16 of core study
UAS7≥16 at Week 12 or Week 16 of core study
Baseline
Observational period[a] (Until UAS7≥16)
Open-label treatment period (NCT04109313)
Follow-up[b]
Remibrutinib 100 mg b.i.d.
Background therapy: second-generation H1-antihistamine[c]
NO
Rescue therapy: Alternative second-generation H1-antihistamine[d]
0–12 weeks
0 4 8 12 16 20 28 40 52 56–68
Visits (Weeks)

[a]Patients who never relapsed (UAS7≥16 at least once) within 12 weeks completed the study at the end of the observational period. [b]The minimum duration of the follow-up period was 4 weeks for all patients who stopped treatment with remibrutinib. Patients who achieved a UAS7 ≤6 at Week 52 of the treatment period extended their follow-up period until they relapsed (UAS7 ≥16). Follow-up ended at Week 68 for all patients. [c]Background therapy was a second-generation H1-AH at a locally approved licensed posology given with a stable treatment regimen. Administration of background H1-AH after Week 4 was at the discretion of the investigator. Background therapy was not permitted from Day 1 until Week 4 of the treatment period. [d]Rescue therapy was a second-generation H1-AH at a locally approved licensed posology that was eliminated primarily via renal excretion. The rescue H1-AH used differed from the background H1-AH and was only given to treat unbearable symptoms (itch) of CSU on a day-to-day basis.

b.i.d., twice daily; H1-AH, H1-antihistamines; N, total number of patients; q.d., once daily; UAS7, weekly Urticaria Activity Score.

RESULTS

- Overall, 194 patients were included in this analysis
- No noticeable CFB (mean±SD) was observed in blood cell count (for leukocytes, neutrophils, eosinophils, basophils, lymphocytes, erythrocytes, and platelets; **Table 1**)

Table 1. Change in hematology parameters

Laboratory parameters	Baseline (N=194)	Week 52 (N=168)	Change from baseline
Leukocytes (x10⁹/L)	6.84±2.14	6.42±1.89	−0.41±1.65
Neutrophils (x10⁹/L)	4.30±1.89	3.85±1.46	−0.47±1.67
Eosinophils (x10⁹/L)	0.181±0.141	0.183±0.133	−0.002±0.131
Basophils (x10⁹/L)	0.046±0.027	0.047±0.026	0.001±0.032
Lymphocytes (x10⁹/L)	1.95±0.62	1.88±0.59	−0.06±0.53
Erythrocytes (x10¹²/L)	4.61±0.42	4.58±0.41[a]	−0.01±0.21
Platelets (x10⁹/L)	263.94±66.35	242.09±59.18[b]	−25.09±45.21

Data are presented as mean±SD. N, total number of patients evaluable. [a]N=171. [b]N=170.

- The blood cell count (mean±SD) remained within the normal limits over 52 weeks of treatment (**Figure 2**)

Figure 2. Blood cell count (mean±SD) for patients treated with remibrutinib

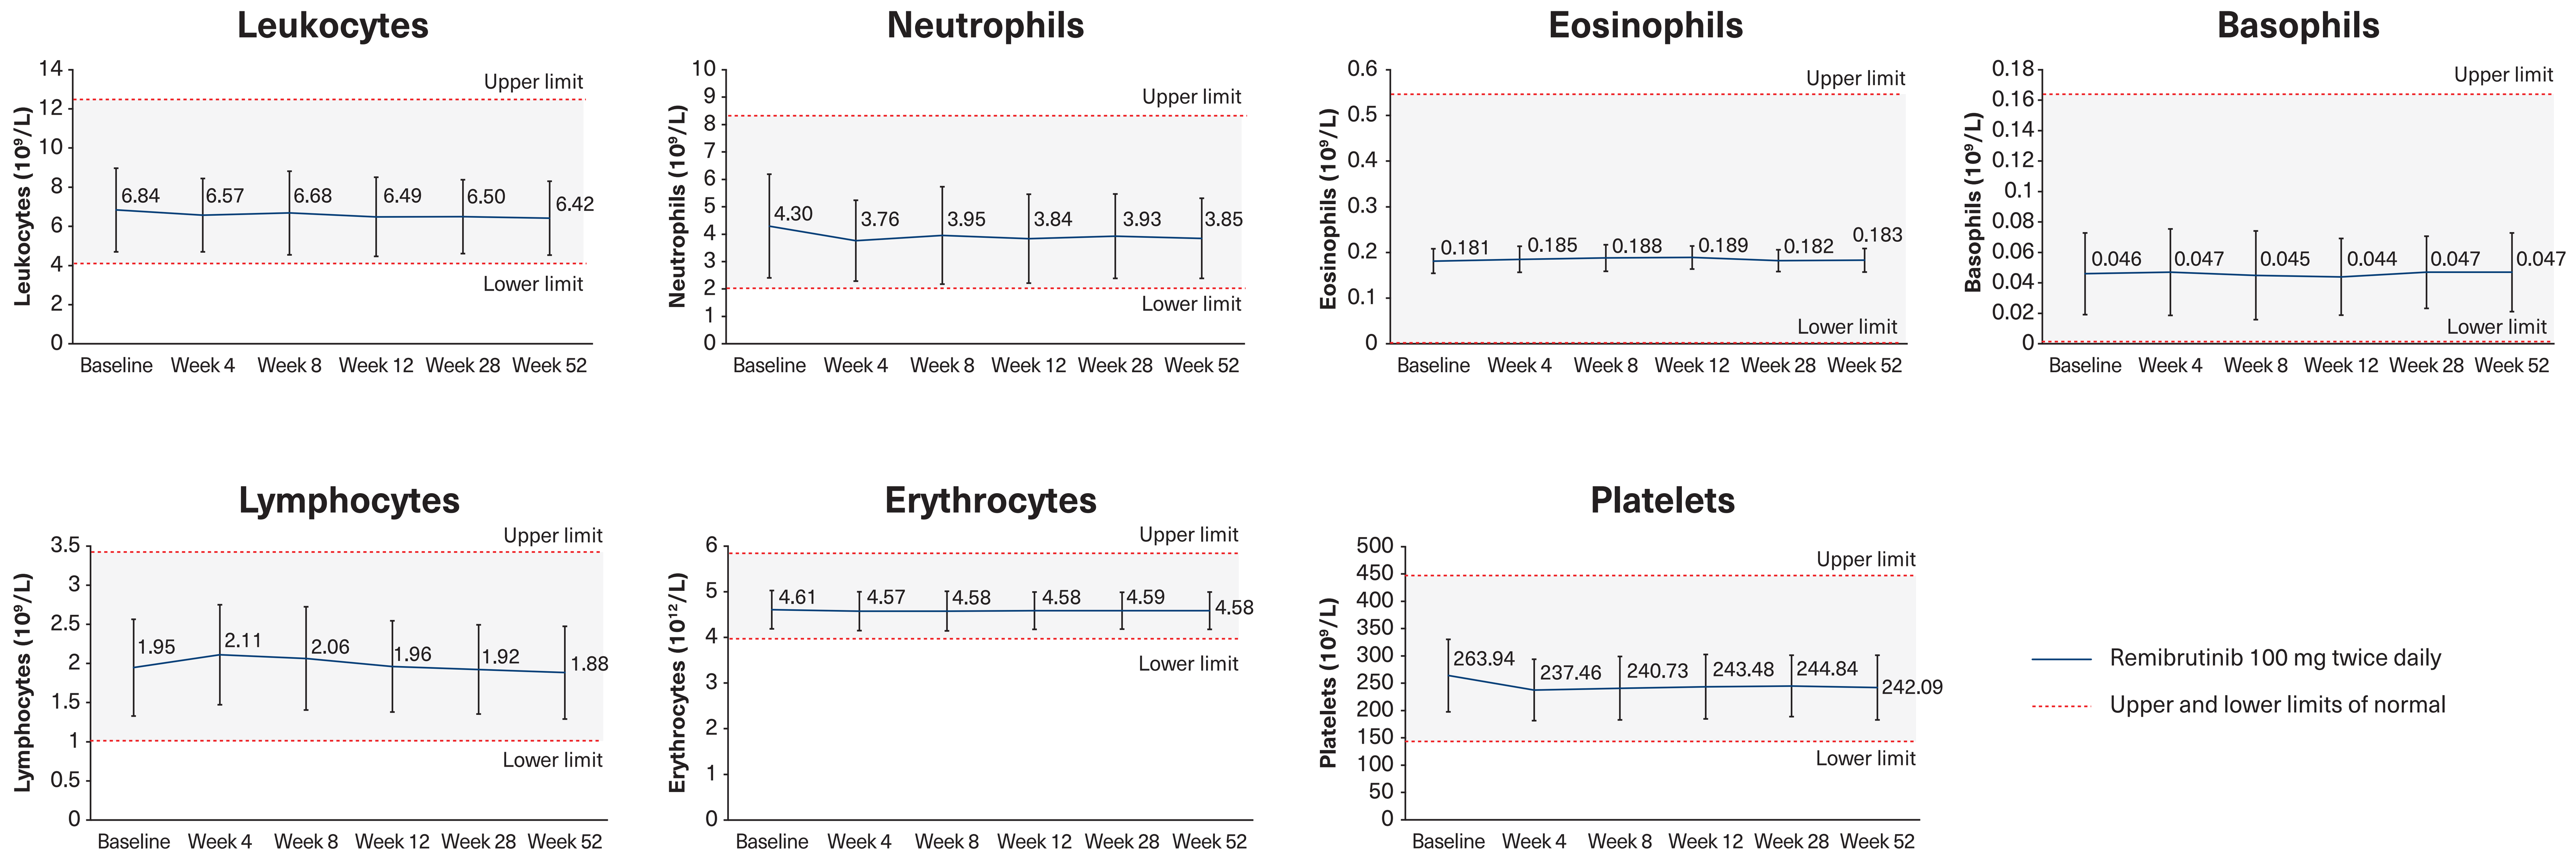
Leukocytes
Leukocytes (10⁹/L)
6.84 6.57 6.68 6.49 6.50 6.42
Neutrophils
Neutrophils (10⁹/L)
4.30 3.76 3.95 3.84 3.93 3.85
Eosinophils
Eosinophils (10⁹/L)
0.181 0.185 0.188 0.189 0.182 0.183
Basophils
Basophils (10⁹/L)
0.046 0.047 0.045 0.044 0.047 0.047
Lymphocytes
Lymphocytes (10⁹/L)
1.95 2.11 2.06 1.96 1.92 1.88
Erythrocytes
Erythrocytes (10¹²/L)
4.61 4.57 4.58 4.58 4.59 4.58
Platelets
Platelets (10⁹/L)
263.94 237.46 240.73 243.48 244.84 242.09
Upper limit
Lower limit
Baseline Week 4 Week 8 Week 12 Week 28 Week 52
Remibrutinib 100 mg twice daily
Upper and lower limits of normal

Error bars represent standard deviation

Disclosures

Alexander Greiner receives grant/research support from Aimmune, Alladapt, ALK, Amphastar, AnaptysBio, Apotex, AstraZeneca, Boehringer Ingelheim, Cephalon, Chiesi, Cipla, Circassia, Clement Clarke, Cour, Cytos, ElectroCore, Genentech, GlaxoSmithKline, Glenmark, Invivodata, Janssen, Kalypsys, Kaz, Lupin, MedImmune, Merck, Mylan, Nestle, Novartis, Ono, Optinose, Pearl, Perrigo, Regeneron, Rigel, Roxane, Sanofi, Schering-Plough, Spirosure (Spirometrix), Sunovion, Teva, UBC/Amgen, Watson and West-Ward, and reports as a consultant to PMI and Novartis. **Vipul Jain** has consulted as and/or advised and/or received research funding from Pediapharm, Medexus, Sanofi, Regeneron, Bausch, Novartis, AbbVie, Aralez, ALK, Celgene, Amgen, LEO Pharma, Mylan, Pfizer, Covis Pharma, Galderma, Eli Lilly and Company, GSK, Kymab, Arcutis Biotherapeutics, and AstraZeneca and also participated as a primary investigator in clinical trials sponsored by Probity medical research Inc, and is a director of Allergy Research Canada Inc. **Ana M Giménez-Arnau** is or recently was a speaker and/or advisor for and/or has received research funding from Almirall, Amgen, AstraZeneca, Avene, Celldex, Escient Pharmaceutials, Genentech, GSK, Instituto Carlos III- FEDER, Leo Pharma, Menarini, Novartis, Sanofi–Regeneron, Servier, Thermo Fisher Scientific, Uriach Pharma / Neucor. **Jeffrey Tillinghast** reports performing clinical research for 3M, Amphastar, AstraZeneca, Cephalon, Genentech, GlaxoSmithKline, Janssen, Lupin, Mylan, Novartis, Roxane, and Teva Pharmaceutical Industries. **Karine Lheritier**, **Sibylle Haemmerle**, and **Artem Zharkov** are employees of Novartis Pharma AG, Basel, Switzerland. **Pauline Walsh** is an employee of Novartis Ireland Limited, Dublin, Ireland. **Robert Snyder** is a speaker and/or advisor for and/or has received research funding from Janssen Biotech, Amgen, Novartis, Galderma, Abbvie, Leo Pharma, Sun Pharma, Regeneron, Sanofi, Mindera, Pfizer, Incyte, Castle Biosciences, and UCB.

Acknowledgments

All authors participated in the development of the poster for presentation. The authors wish to thank all investigators and patients involved in the trial. The authors thank **Sagar Wagh** and **Mohammad Fahad Haroon** for editorial and medical writing support, and **Rupa De** for the design support (all Novartis Healthcare Pvt. Ltd., Hyderabad, India), which was funded by Novartis Pharma AG, Basel, Switzerland, in accordance with the Good Publication Practice (GPP3) guidelines (http://www.ismpp.org/gpp3).

Funding

This study was sponsored by Novartis Pharma AG, Basel, Switzerland.

References

1. Zuberbier T, et al. *Allergy*. 2022;77(3):734–766.
2. Angst D, et al. *J Med Chem*. 2020;63:5102–5118.
3. Maurer M, et al. *J Allergy Clin Immunol*. 2022;150(6):1498–1506.e2.
4. Jain V, et al. *J Allergy Clin Immunol*. 2023;S0091-6749(23)01346-5. doi: 10.1016/j.jaci.2023.10.007. Epub ahead of print.
5. Saini S, et al. *ACAAI 2023*. Oral Presentation LB001 – Late-breaker; November 12, 2023; Anaheim, CA.
6. Xia S, et al. *Cell Mol Immunol*. 2020;17(10):1042–1052.